A mechanistic model for prediction of metastatic relapse in early-stage breast cancer using routine clinical features

Célestin Bigarré, Pascal Finetti, François Bertucci, Xavier Muracciole, Sébastien Benzekry

To cite this version:

Célestin Bigarré, Pascal Finetti, François Bertucci, Xavier Muracciole, Sébastien Benzekry. A mechanistic model for prediction of metastatic relapse in early-stage breast cancer using routine clinical features. AACR, Apr 2022, New Orléans, United States. pp.2737, 10.1158/1538-7445.AM2022-2737 . hal-03752407

HAL Id: hal-03752407
https://inria.hal.science/hal-03752407

Submitted on 16 Aug 2022

HAL is a multi-disciplinary open access archive for the deposit and dissemination of scientific research documents, whether they are published or not. The documents may come from teaching and research institutions in France or abroad, or from public or private research centers.

L'archive ouverte pluridisciplinaire **HAL**, est destinée au dépôt et à la diffusion de documents scientifiques de niveau recherche, publiés ou non, émanant des établissements d'enseignement et de recherche français ou étrangers, des laboratoires publics ou privés.

Mechanistic modeling of metastatic free relapse in breast cancer to investigate the biological impact of diagnosis biomarkers.

Nº 4426

Célestin BIGARRÉ [1], Pascal FINETTI[2], François BERTUCCI[2], Xavier MURACCIOLE[1,3], Sébastien BENZEKRY [1]

[1] COMPutational pharmacology and clinical Oncology Department, Cancer Research Center of Marseille (CRCM), Inserm U1068, CNRS UMR7258, Institut Paoli-Calmettes, Inria Sophia Antipolis – Méditerranée, Marseille, France

[2] Predictive Oncology Laboratory, Cancer Research Center of Marseille (CRCM), INSERM, U1068, CNRS, UMR7258, Institut Paoli-Calmettes, Aix Marseille Université, Marseille, France

[3] Radiotherapy Department, Assistance Publique-Hôpitaux de Marseille, Aix Marseille University, Marseille, France

BACKGROUND

◆ Prediction of metastasis is a major challenge for treatment individualization of breast cancer patients.

◆ The time to metastatic relapse in breast cancers has been studied mainly with classical statistical models (e.g. Cox proportional hazards model).

◆ Mechanistic models can be used not only for prediction but also to explore the biological role of predictive variables.

OBJECTIVES

◆ Describe and predict metastasis free survival in a population of breast cancer patients who did not receive systemic adjuvant therapies.

◆ Use mechanistic modeling to disentangle the biological roles of biomarkers between growth and dissemination

MATERIAL AND METHODS

Mechanistic model of the metastatic relapse[1]

Tumor growth (Gompertz): $\frac{\mathrm{d}V}{\mathrm{d}t} = (\alpha - b\log V)V$

Dissemination rate: $d(V_p) = \mu V_p$

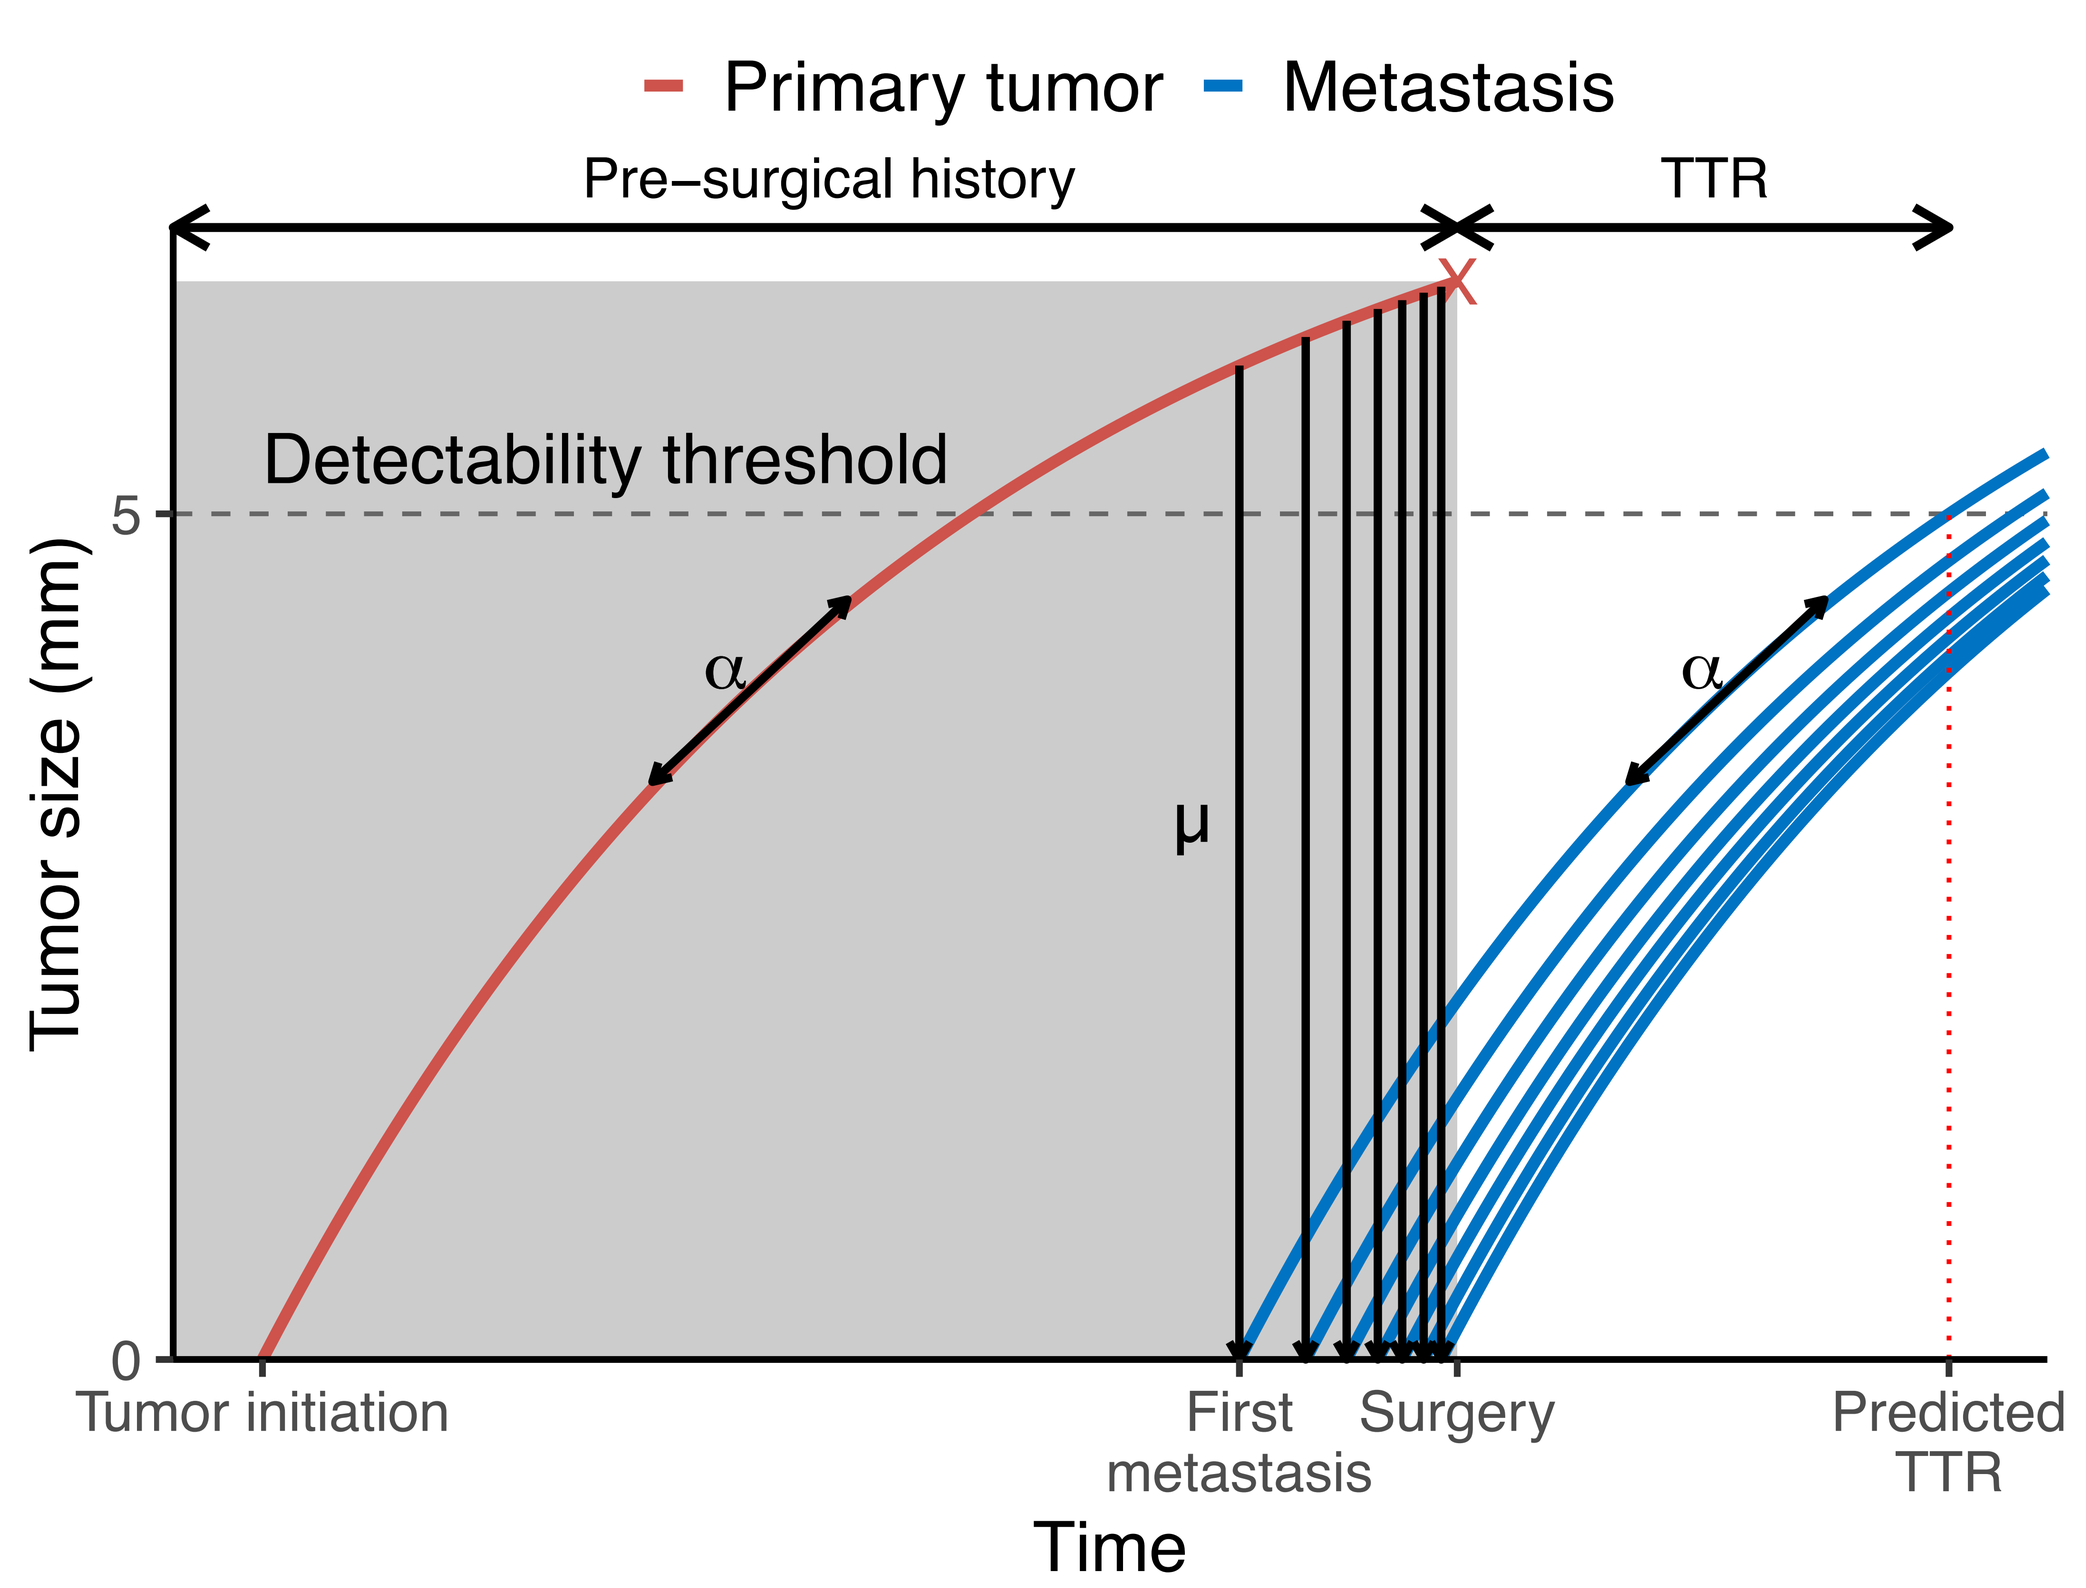
Primary tumor
Metastasis
Pre–surgical history
TTR
Detectability threshold
Tumor size (mm)
α
μ
Tumor initiation
First metastasis
Surgery
Predicted TTR
Time

Mixed-effects model

$$T^i = TTR(V^i_{diag}, \alpha^i, \mu^i) + \mathcal{N}(0, \varepsilon^2)$$

$$\begin{cases} \log \alpha^i = \log \alpha_{pop} + {}^T\beta_\alpha \cdot C^i + \mathcal{N}(0, \omega_\alpha^2) \\ \log \mu^i = \log \mu_{pop} + {}^T\beta_\mu \cdot C^i + \mathcal{N}(0, \omega_\mu^2) \end{cases}$$

◆ The population parameters of the models were estimated with the SAEM algorithm.

◆ Covariates significance was assessed by univariate bootstraps.

◆ The final covariate model structures were selected by a backward elimination procedure on the BIC.

Data

591 patients treated at the Bergonié Institute (Bordeaux, France), 676 patients from public databases aggregated by the Paoli Calmettes Institute (IPC, Marseille, France), with routine clinical features (tumor size at diagnosis, age, grade, molecular subtype, TNM, ER, PR, number of invaded nodes, HER2, Ki67) and 163 patients from AP-HM with routine clinical features as well as three other biomarkers (Urokinase Plasminogen Activator, Plasminogen Activator Inhibitor-1, Thymidine Kinase-1).

RESULTS

Model fits

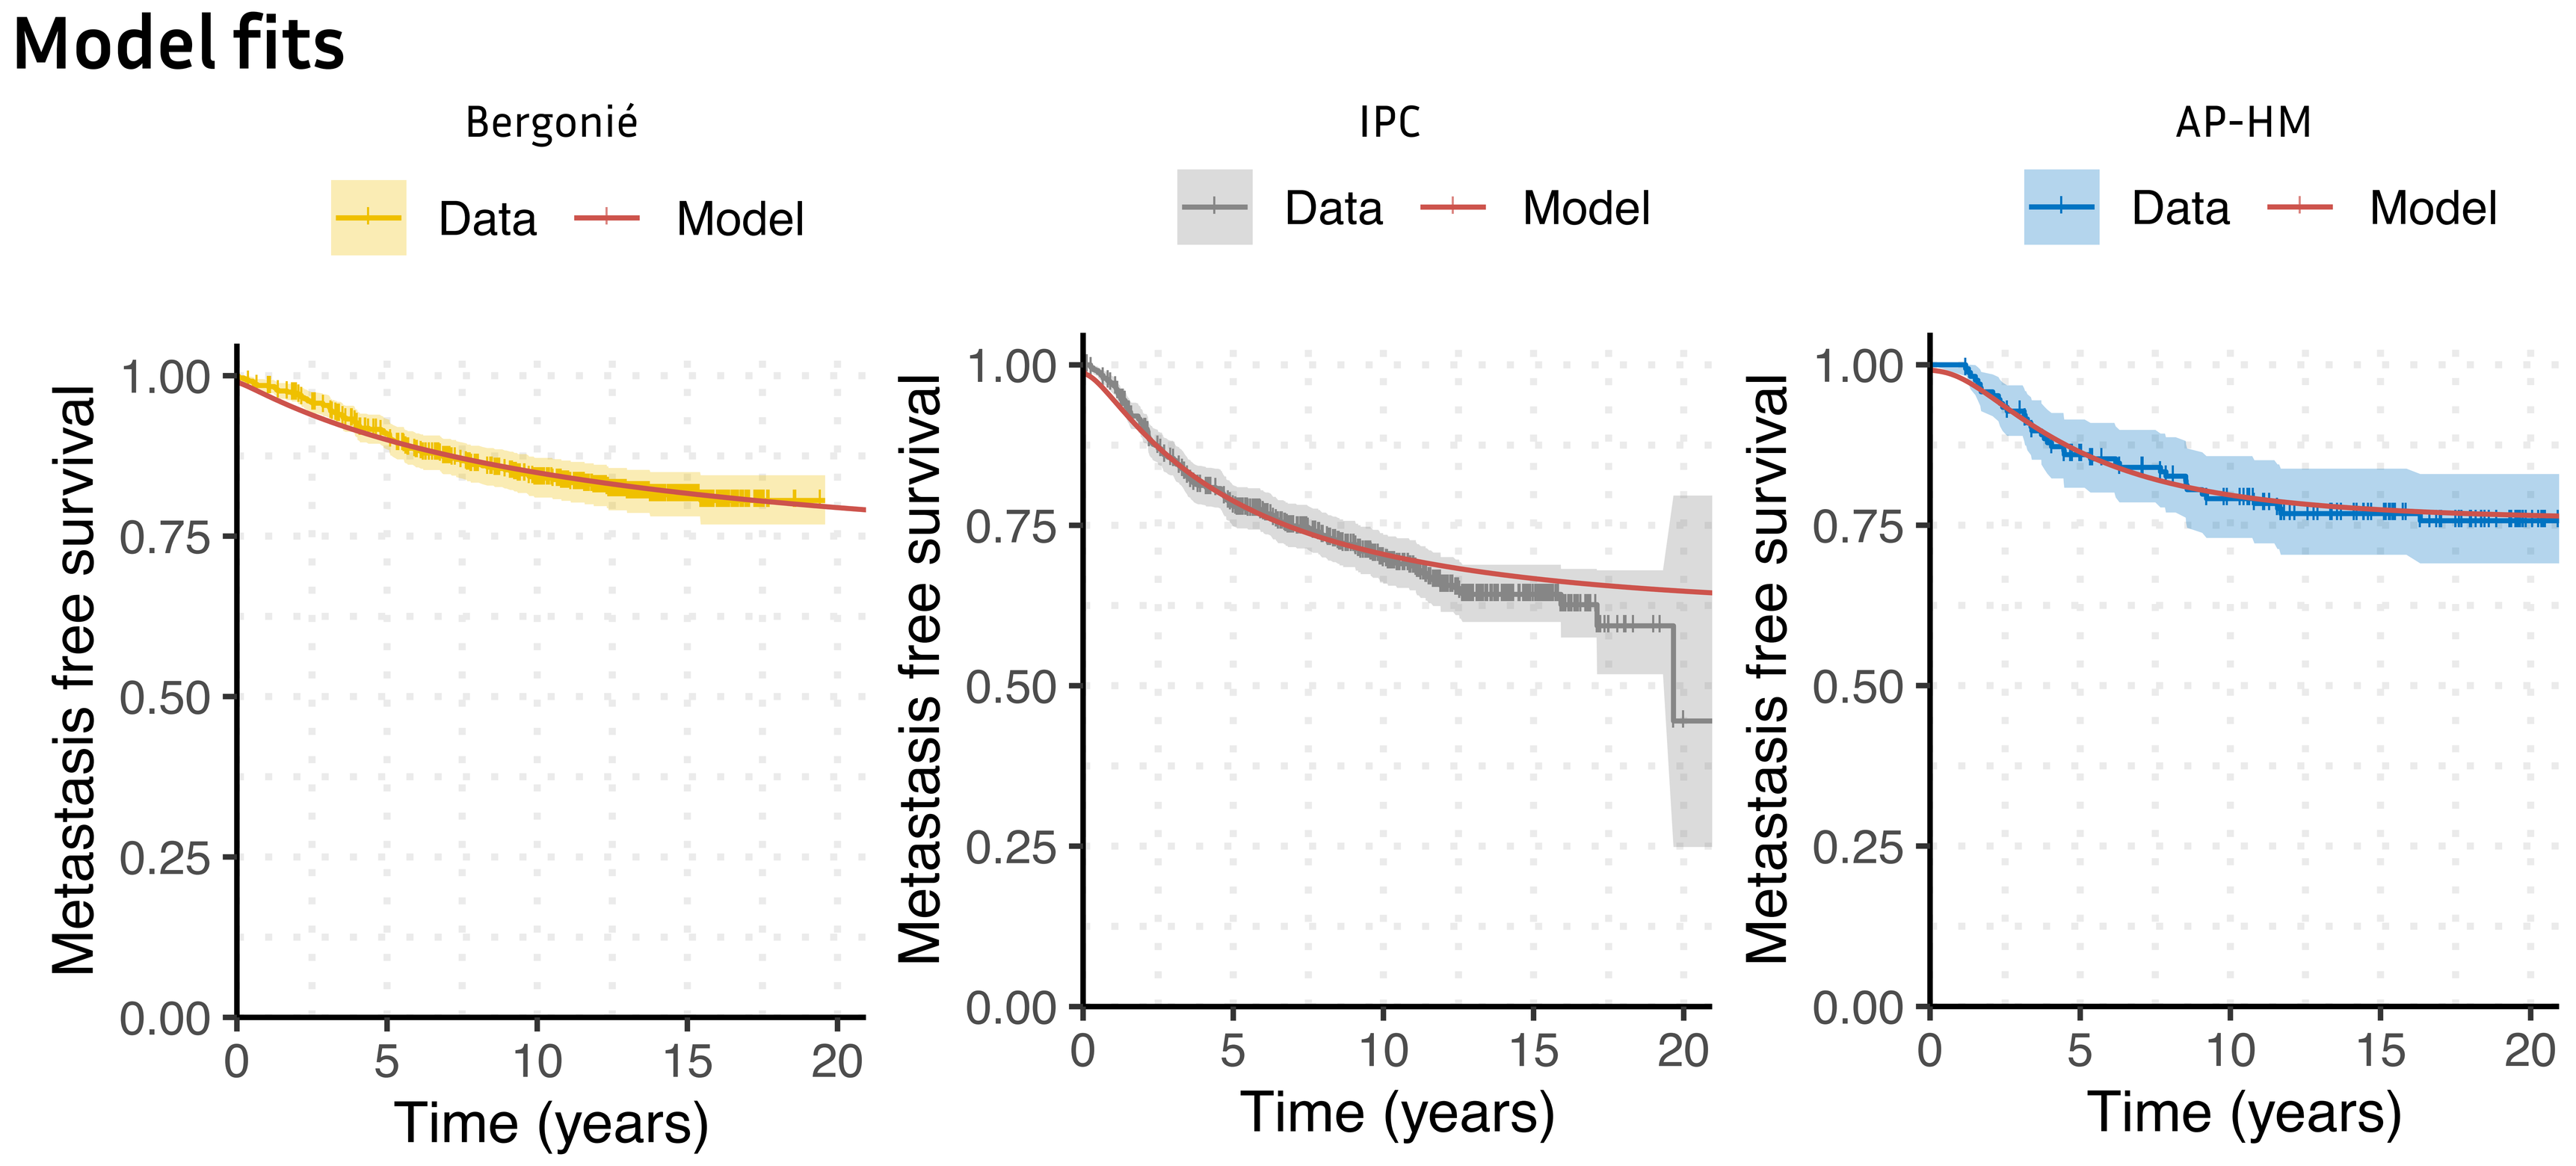
Bergonié
IPC
AP-HM
Data
Model
Metastasis free survival
Time (years)

Kaplan-Meier of the Metastasis free survival for all three databases and predictions of the corresponding models without covariates effects other than the size of the tumor (structurally present).

Estimation of the parameters

	Bergonié		IPC		AP-HM
$\log \alpha_{pop}$	-5.3 (25.4%)		-5.28 (4.43%)		-4.3 (9.85%)
$\log \mu_{pop}$	-29.7 (1.77%)		-29.2 (0.637%)		-32 (2.89%)
σ	0.571 (86.4%)		0.511 (48.6%)		0.0364 (251%)
ω_α	1.05 (257%)		0.734 (36.3%)		0.722 (10.8%)
ω_μ	4.94 (1.69%)		4.2 (9.06%)		3.27 (9.74%)
$\beta_{\alpha, \mathrm{ER}}$	-0.99 (53.2%)	$\beta_{\alpha, \mathrm{grade\ 2}}$	0.61 (43.7%)		
$\beta_{\alpha, \mathrm{Ki67}}$	1.38 (35.8%)	$\beta_{\alpha, \mathrm{grade\ 3}}$	1.67 (14.8%)		
		$\beta_{\alpha, \mathrm{PR}}$	-0.549 (34%)		
		$\beta_{\mu, \mathrm{HER2}}$	1.99 (31.6%)	$\beta_{\mu, \mathrm{PAI\text{-}1}}$	0.30 (30.2%)
		$\beta_{\mu, \mathrm{nodes}}$	2.72 (47.4%)		

Values of the parameters for the selected models, estimation (relative standard error %). Estimation was performed with the SAEM algorithm, standard deviation were obtained by bootstrap.

Variable selection

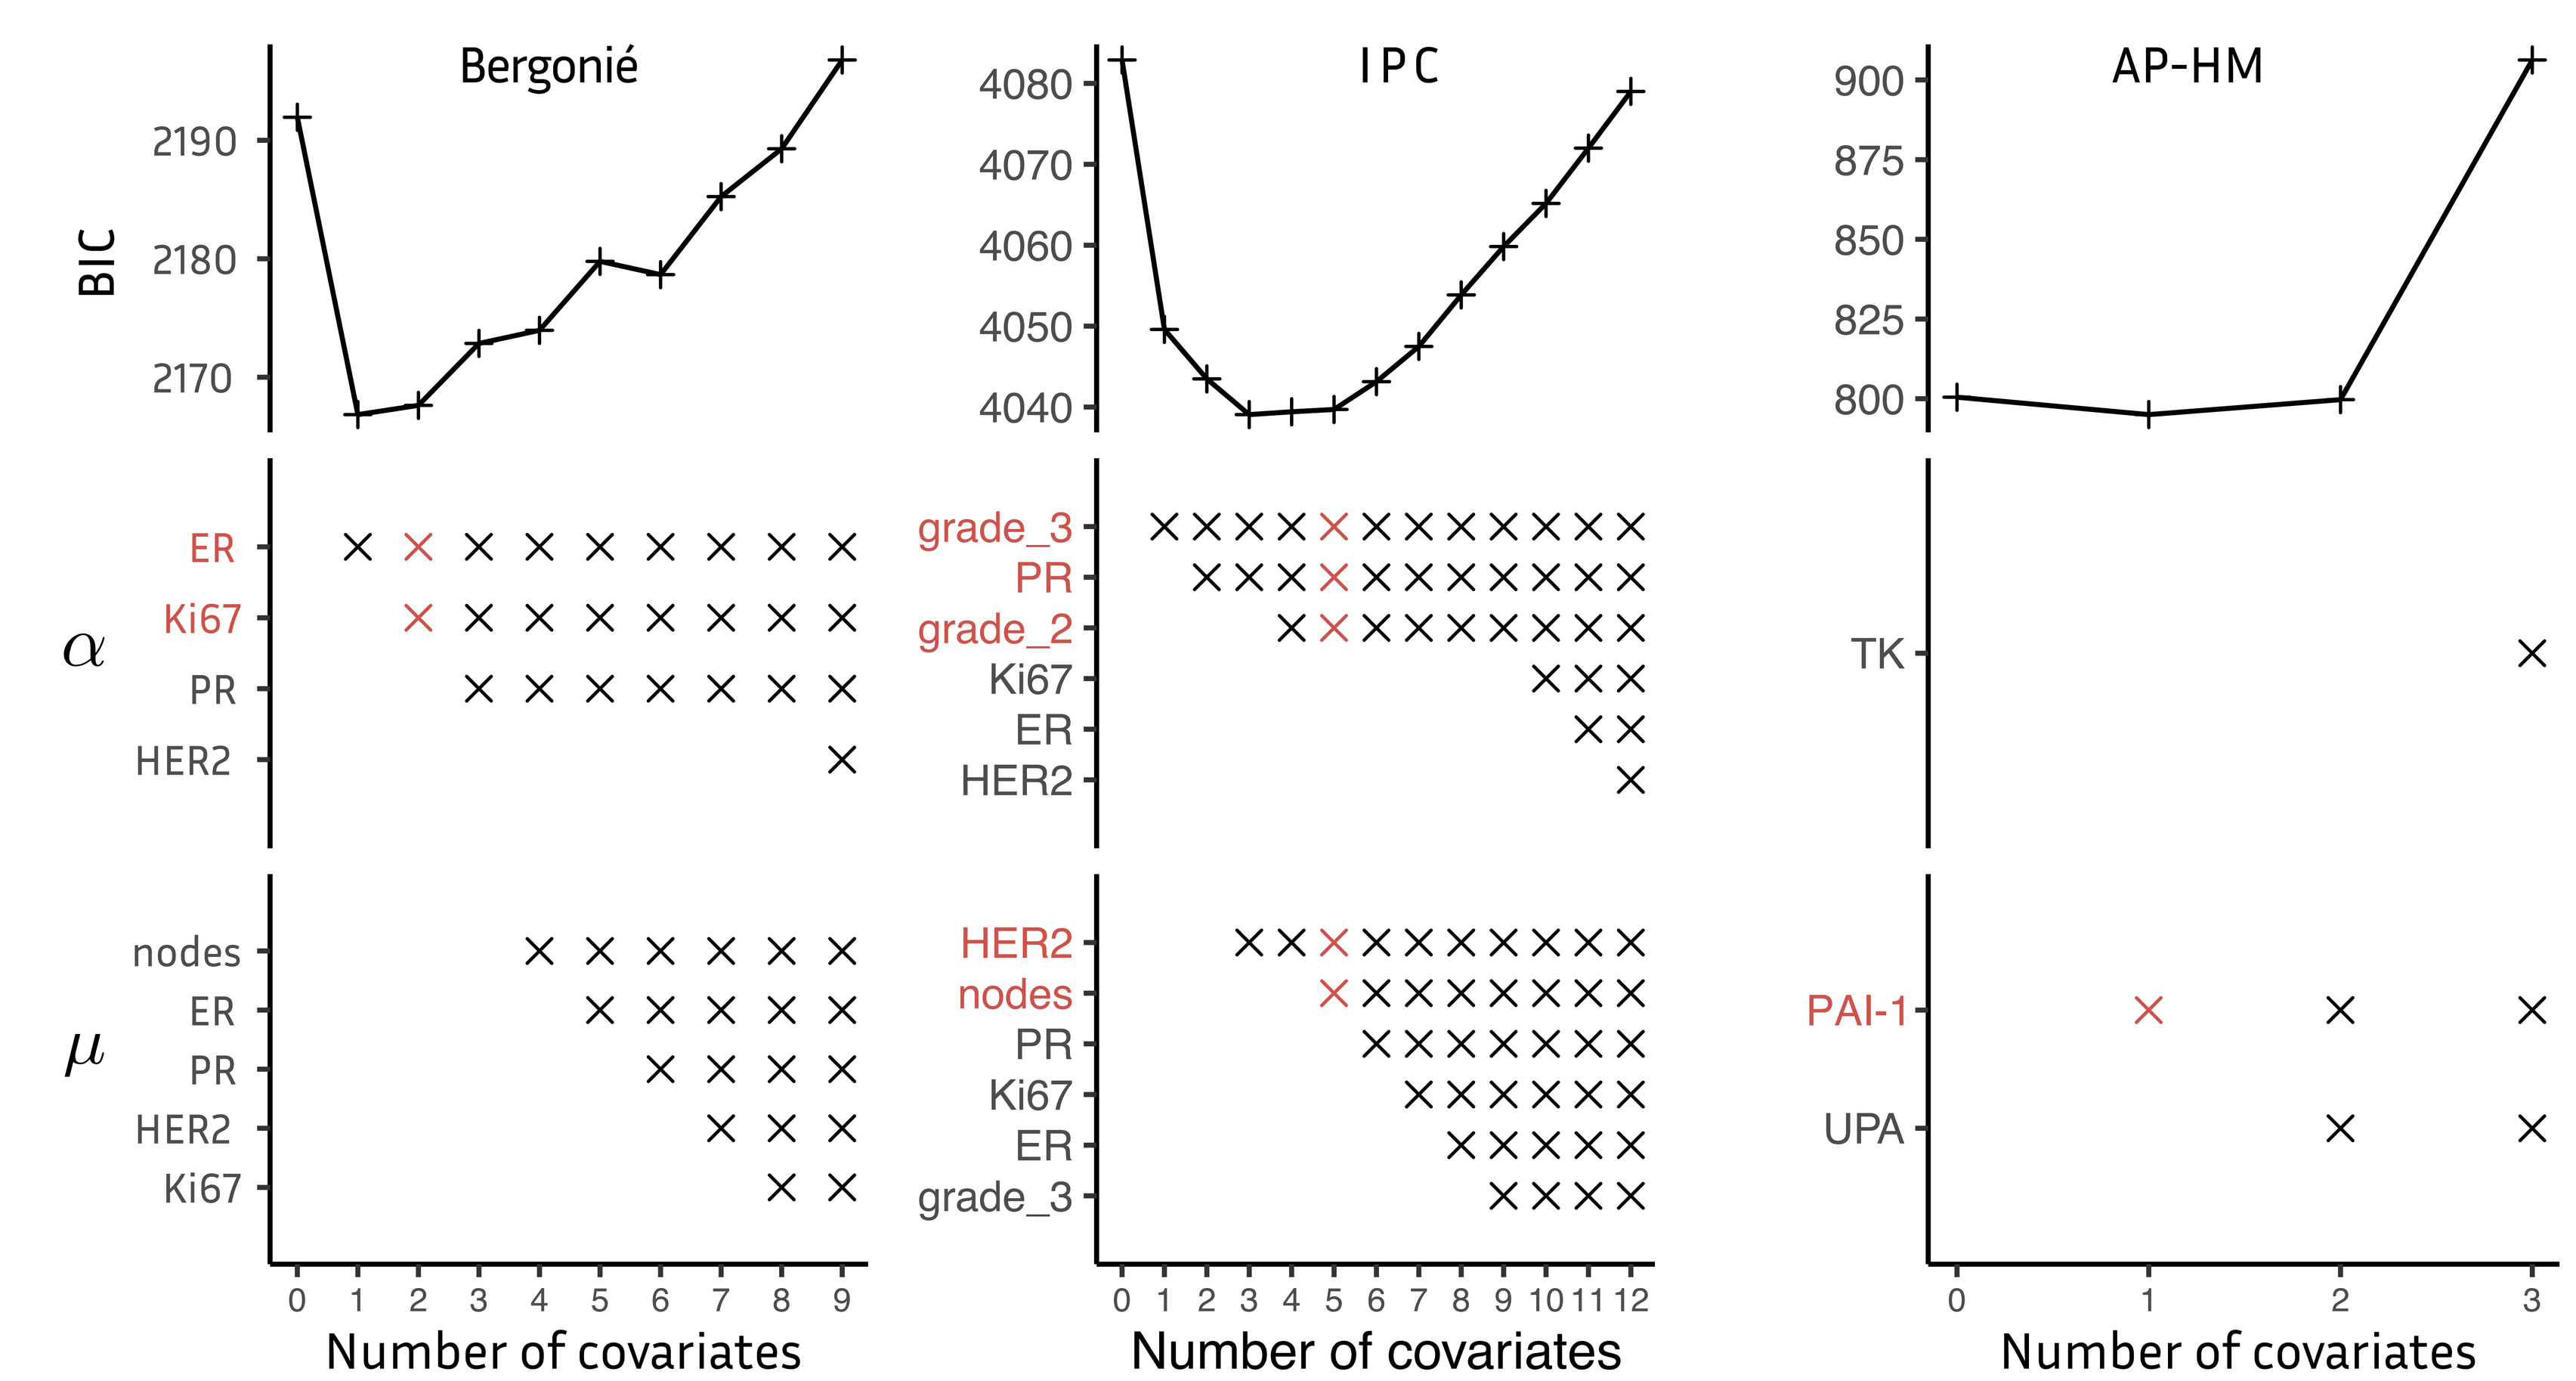
Bergonié
IPC
AP-HM
BIC
α
μ
ER
Ki67
PR
HER2
nodes
grade_3
grade_2
TK
PAI-1
UPA
Number of covariates

Backward procedure on BIC the for model selection, selected models are highlighted in red. The variables presented were selected as significant in a univariate bootstrap analysis.

Calibration and prediction

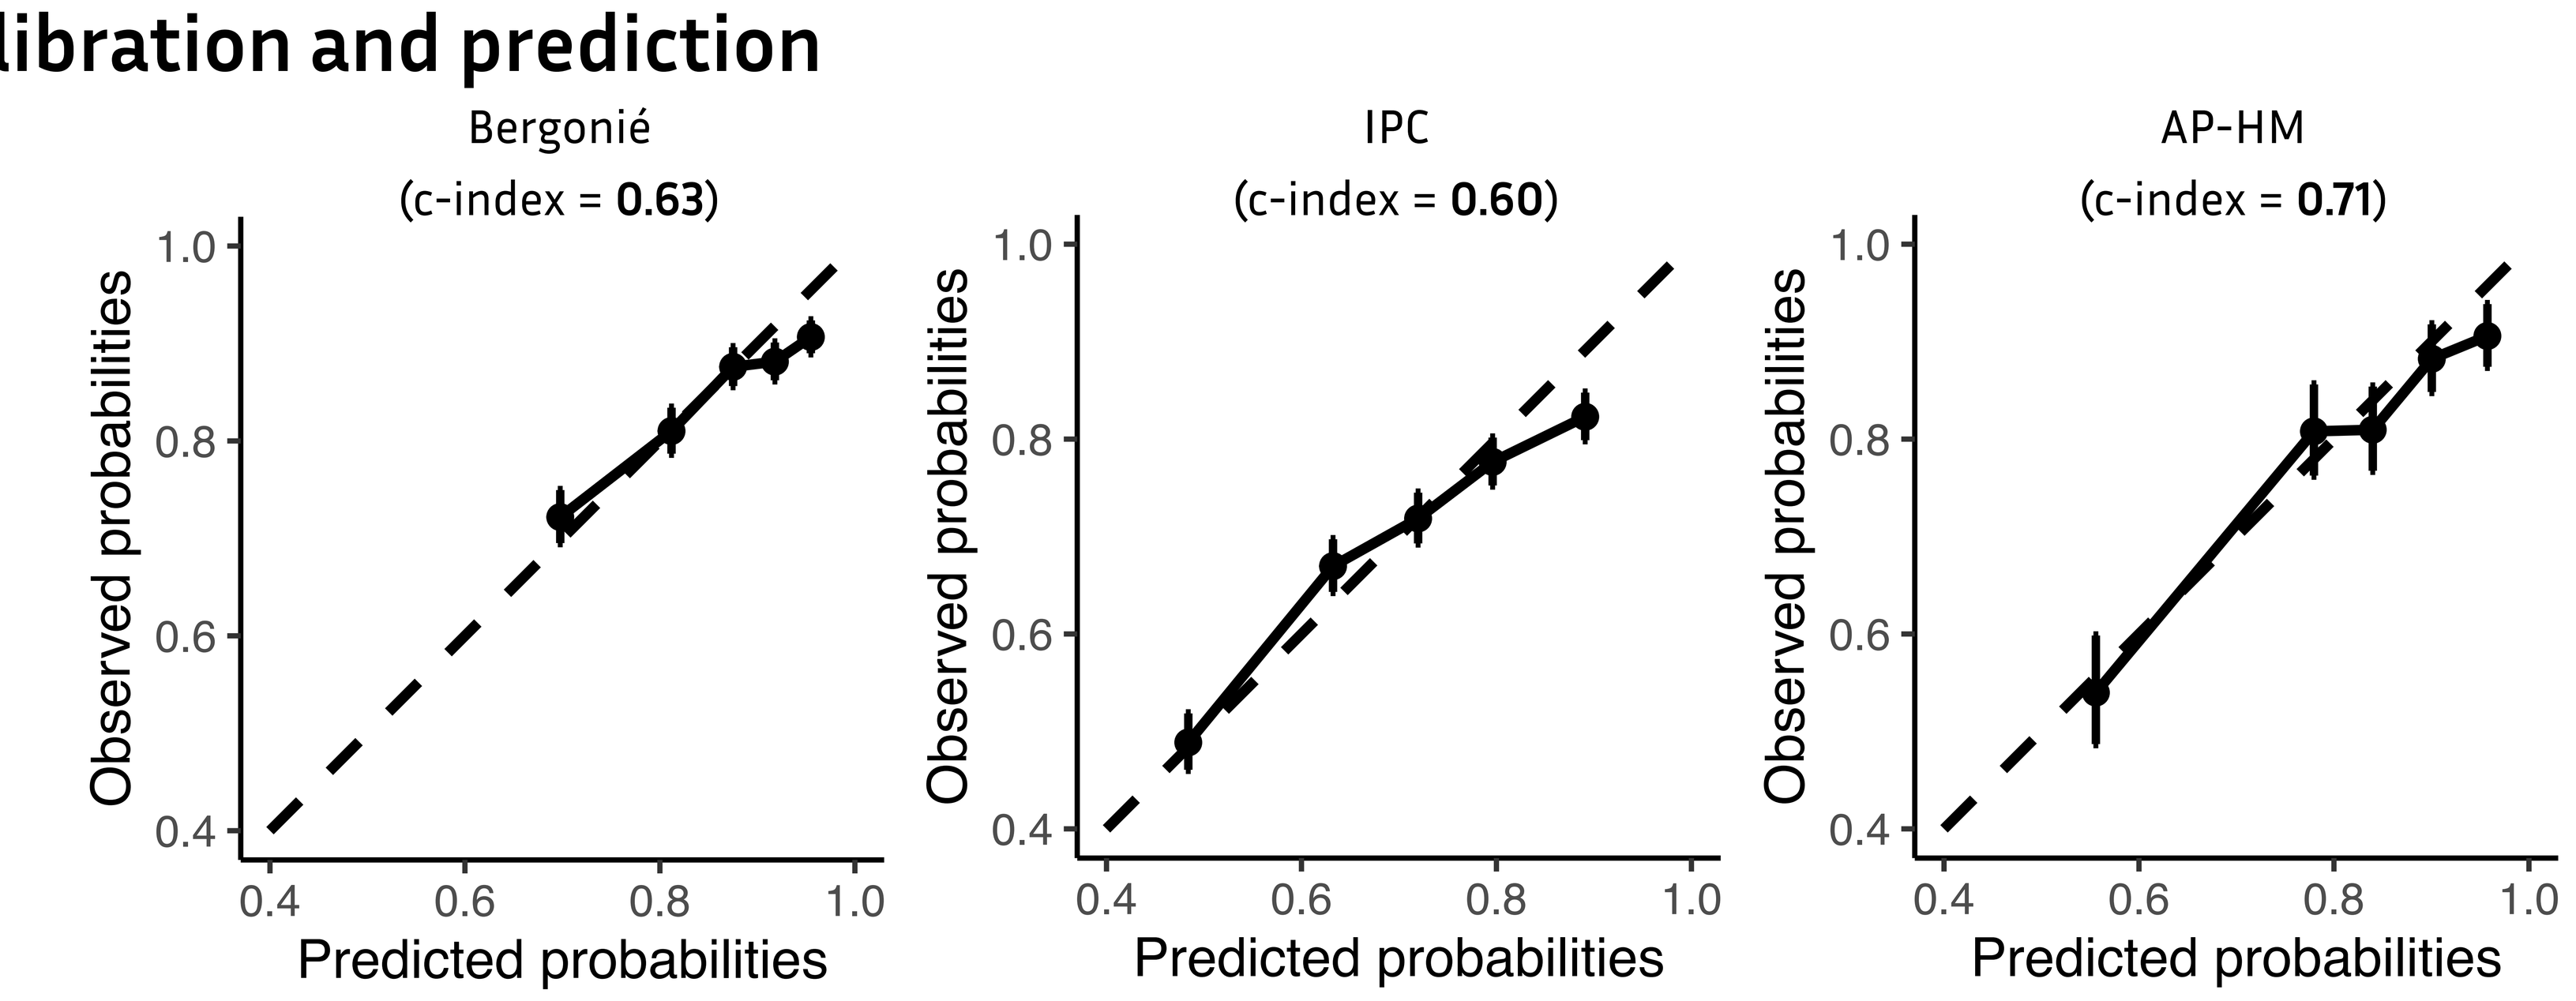
Bergonié
(c-index = **0.63**)
IPC
(c-index = **0.60**)
AP-HM
(c-index = **0.71**)
Observed probabilities
Predicted probabilities

Calibration at 10 years of the selected models.

CONCLUSIONS AND PERSPECTIVES

◆ Using mechanistic model of the metastatic relapse, we can adequately describe the metastasis free survival in our study population

◆ Predictive performances were excellent in calibration but mitigated in discrimination.

◆ The model associates HER2, nodal status and PAI-1 with the metastatic dissemination, and TK, Ki67 and grade with tumor growth. These finding are concordant with the known biological roles of the biomarkers.

References

[1] *Nicolò et al. (Benzekry), Machine Learning and Mechanistic Modeling for Prediction of Metastatic Relapse in Early-Stage Breast Cancer. Jco Clin Cancer Informatics 4, 259–274 (2020).*